In vivo engineered and lineage-restricted multiplexed CAR-M, -NK, and -T cell therapy mounts robust solid tumor control in pre-clinical models

Yiwen Zhao, Chirayu Chokshi, Alvin Pratama, Dhruv Varshney, Dennis Ramos Trinidad, Cristina Santoriello, Ying-Cing Lin, Charles Fox, Alexey Seregin, Adam Golding, Chapman Wright, Yinghua Wang, Joe Salas, and Corinne Decker
Ensoma, Inc. Boston, MA USA

Abstract

Background *Ex vivo* chimeric antigen receptor (CAR)-T cell therapy has achieved remarkable clinical success in hematologic malignancies. Such transformative efficacy, however, has yet to be realized in solid tumors where CAR-T activity is hindered by poor trafficking and infiltration, the immunosuppressive tumor microenvironment (TME), and limited persistence. CAR-myeloid cells (CAR-M) can infiltrate and modulate the TME, enhancing T cell activity, but their short persistence limits clinical efficacy. Complex manufacturing, high cost, and need for lymphodepletion present additional obstacles that limit patient accessibility to *ex vivo* cell therapy. To address these challenges, we are pursuing an *in vivo* multicellular CAR therapy to enable durable and synergistic anti-tumor activity in solid tumors. **Methods** We developed a virus-like particle (VLP) platform using an optimized helper-dependent adenovirus that preferentially targets long-term hematopoietic stem cells (LT-HSCs) via CD46 and harbors a large 35-kilobase cargo capacity. Mobilization of HSCs into the blood enables *in situ* transduction. Co-transduction of the therapeutic payload with a VLP harboring Sleeping Beauty transposase facilitates stable transgene integration in HSCs. **Results** Lineage-specific HER2 CAR expression was validated *in vitro* in both human and murine primary cells. Mice expressing human HER2 (huHER2 mice) were transplanted with HSPCs transduced with VLPs encoding lineage-restricted HER2 CARs. Following enrichment, HSC-derived HER2 CAR-expressing monocytes, NK cells, and T cells were observed in peripheral blood. HER2 CAR significantly controlled the tumor growth of human HER2-expressing EO771 mammary fat tumors and showed significant survival benefit. No health issues occurred in the huHER2 mice during the period of tumor control, and body weight remained stable for the period of observation for over two months. **Conclusions** Here, we establish a robust platform for *in vivo* HSC-derived multicellular CAR immune cell engineering that combines the tumor remodeling potential of CAR-M with the potent cytotoxicity of CAR-T and -NK cells in a single therapeutic payload administered in one *in vivo* dose. Unlike other *in vivo* cell therapy platforms that target mature immune cells, engineering at the HSC level generates a potentially long-lived, self-renewing pool of CAR-expressing effectors. Collectively, this technology delivering a multiplexed CAR-M, -NK, and -T cell payload offers an off-the-shelf therapy with the potential to mediate long-term anti-tumor activity.

Engenious™ Platform for *in vivo* gene delivery

Virus-like particle (VLP)

LT-HSC targeting
Ad35++ fiber targets CD46 on HSCs

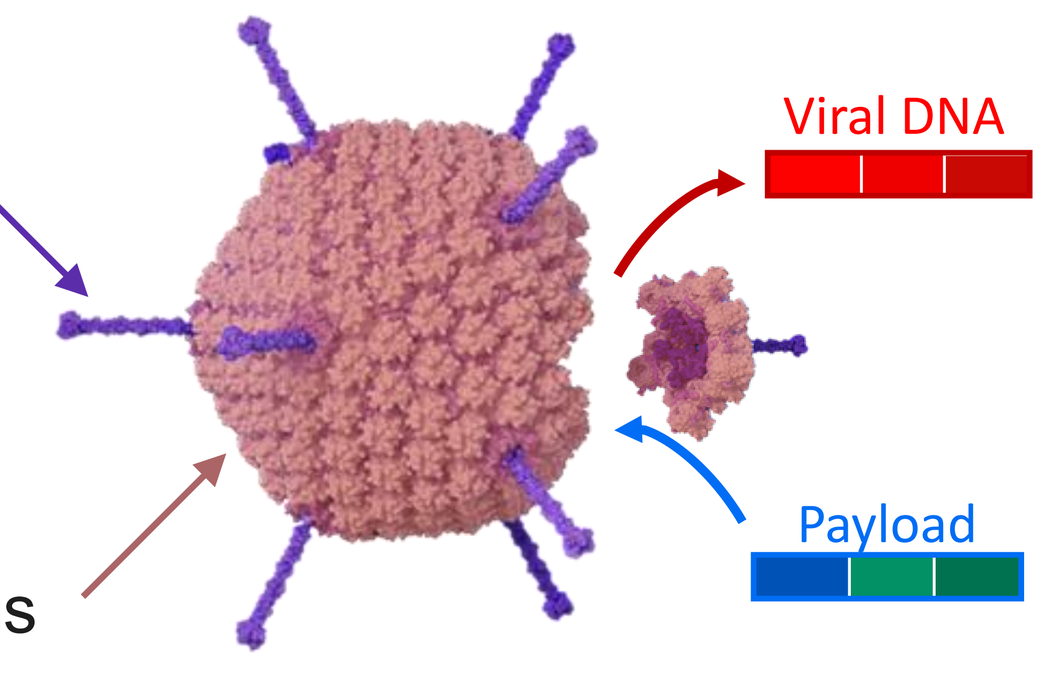
Viral DNA
Payload

"Gutless" vector
Contains no viral genes

Delivery to nucleus
Ad5 capsid shuttles DNA to cell nucleus

35 kB payload capacity
Enables multiplexed gene insertion

Multi-lineage CAR strategy for solid tumors

HSC engineering generates a ***self-renewing, multi-cellular*** therapy

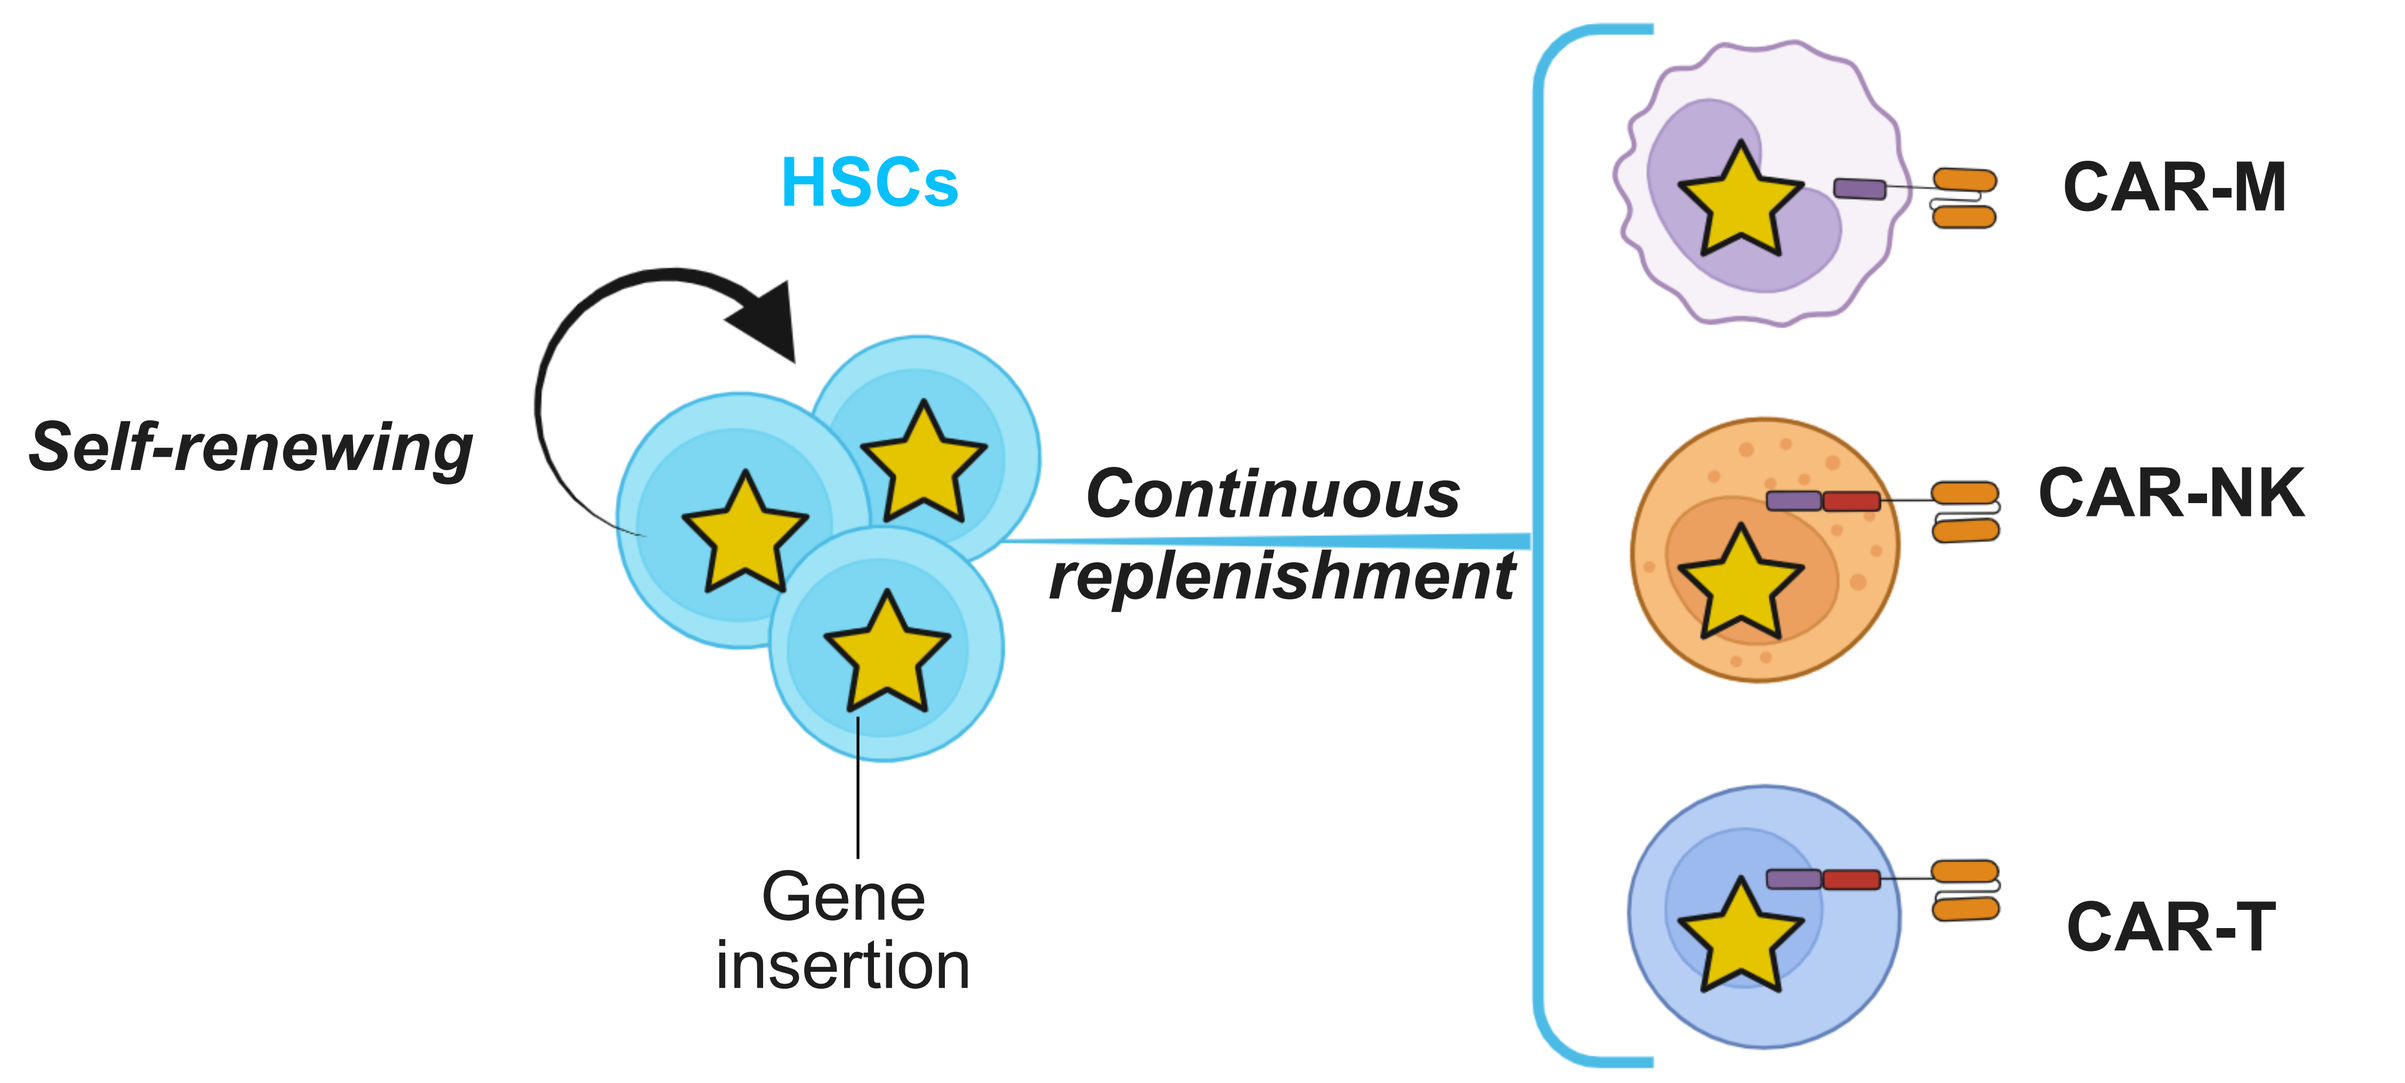
HSCs
Self-renewing
Continuous replenishment
Gene insertion
CAR-M
CAR-NK
CAR-T

Multiplexed, lineage-restricted vector design to generate HER2 CAR-M, NK, & T cells from a single transgene

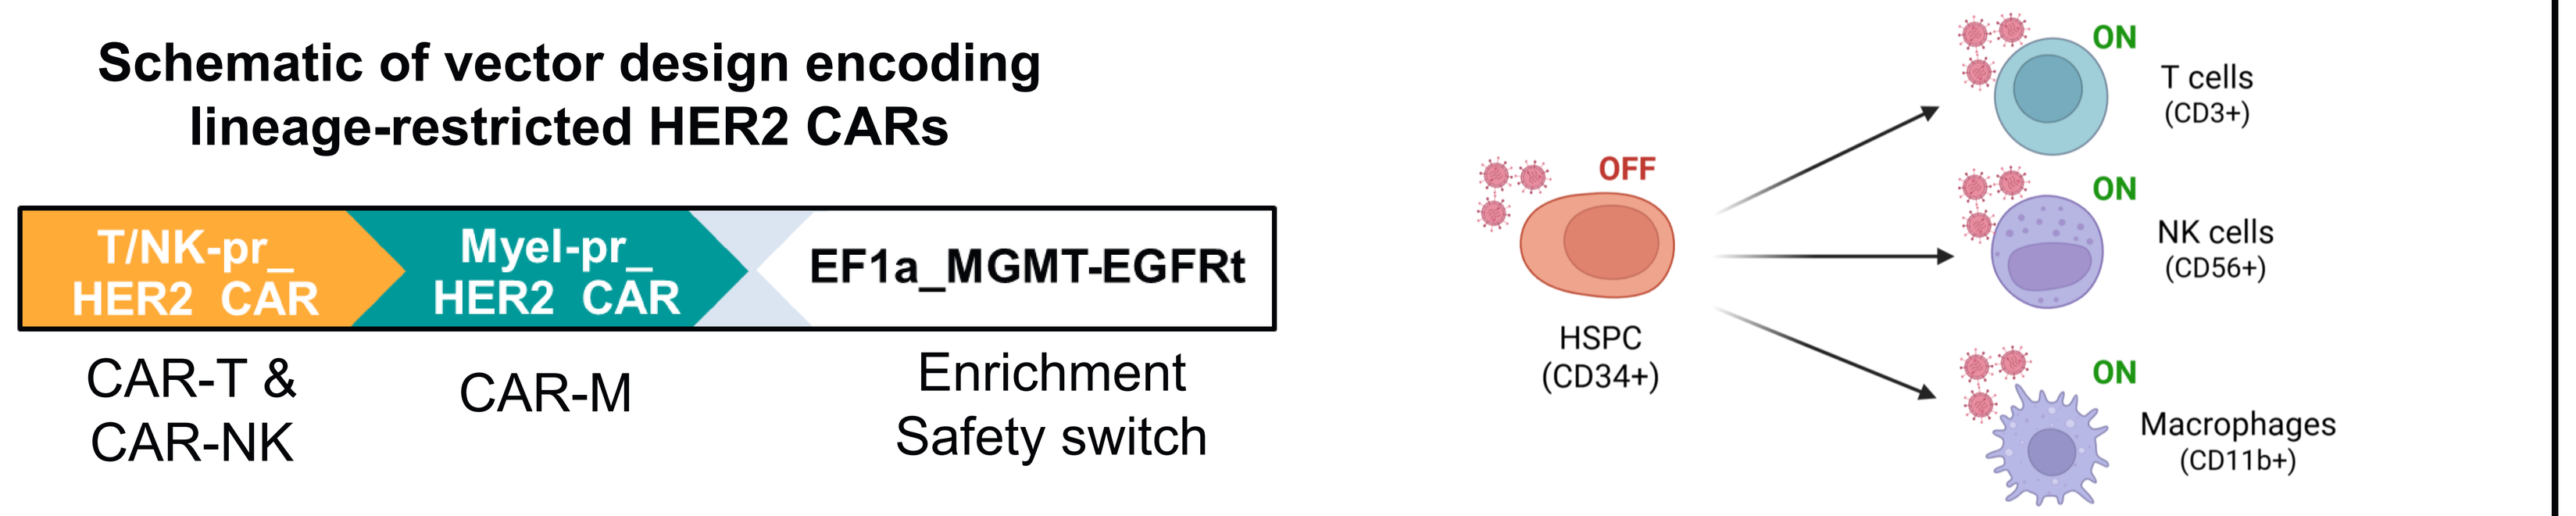
Schematic of vector design encoding lineage-restricted HER2 CARs
T/NK-pr_ HER2 CAR
Myel-pr_ HER2 CAR
EF1a_MGMT-EGFRt
CAR-T & CAR-NK
CAR-M
Enrichment Safety switch
OFF
HSPC (CD34+)
ON
T cells (CD3+)
ON
NK cells (CD56+)
ON
Macrophages (CD11b+)

Lineage-specific promoters drive functional HER2 CAR expression in human immune cells *in vitro*

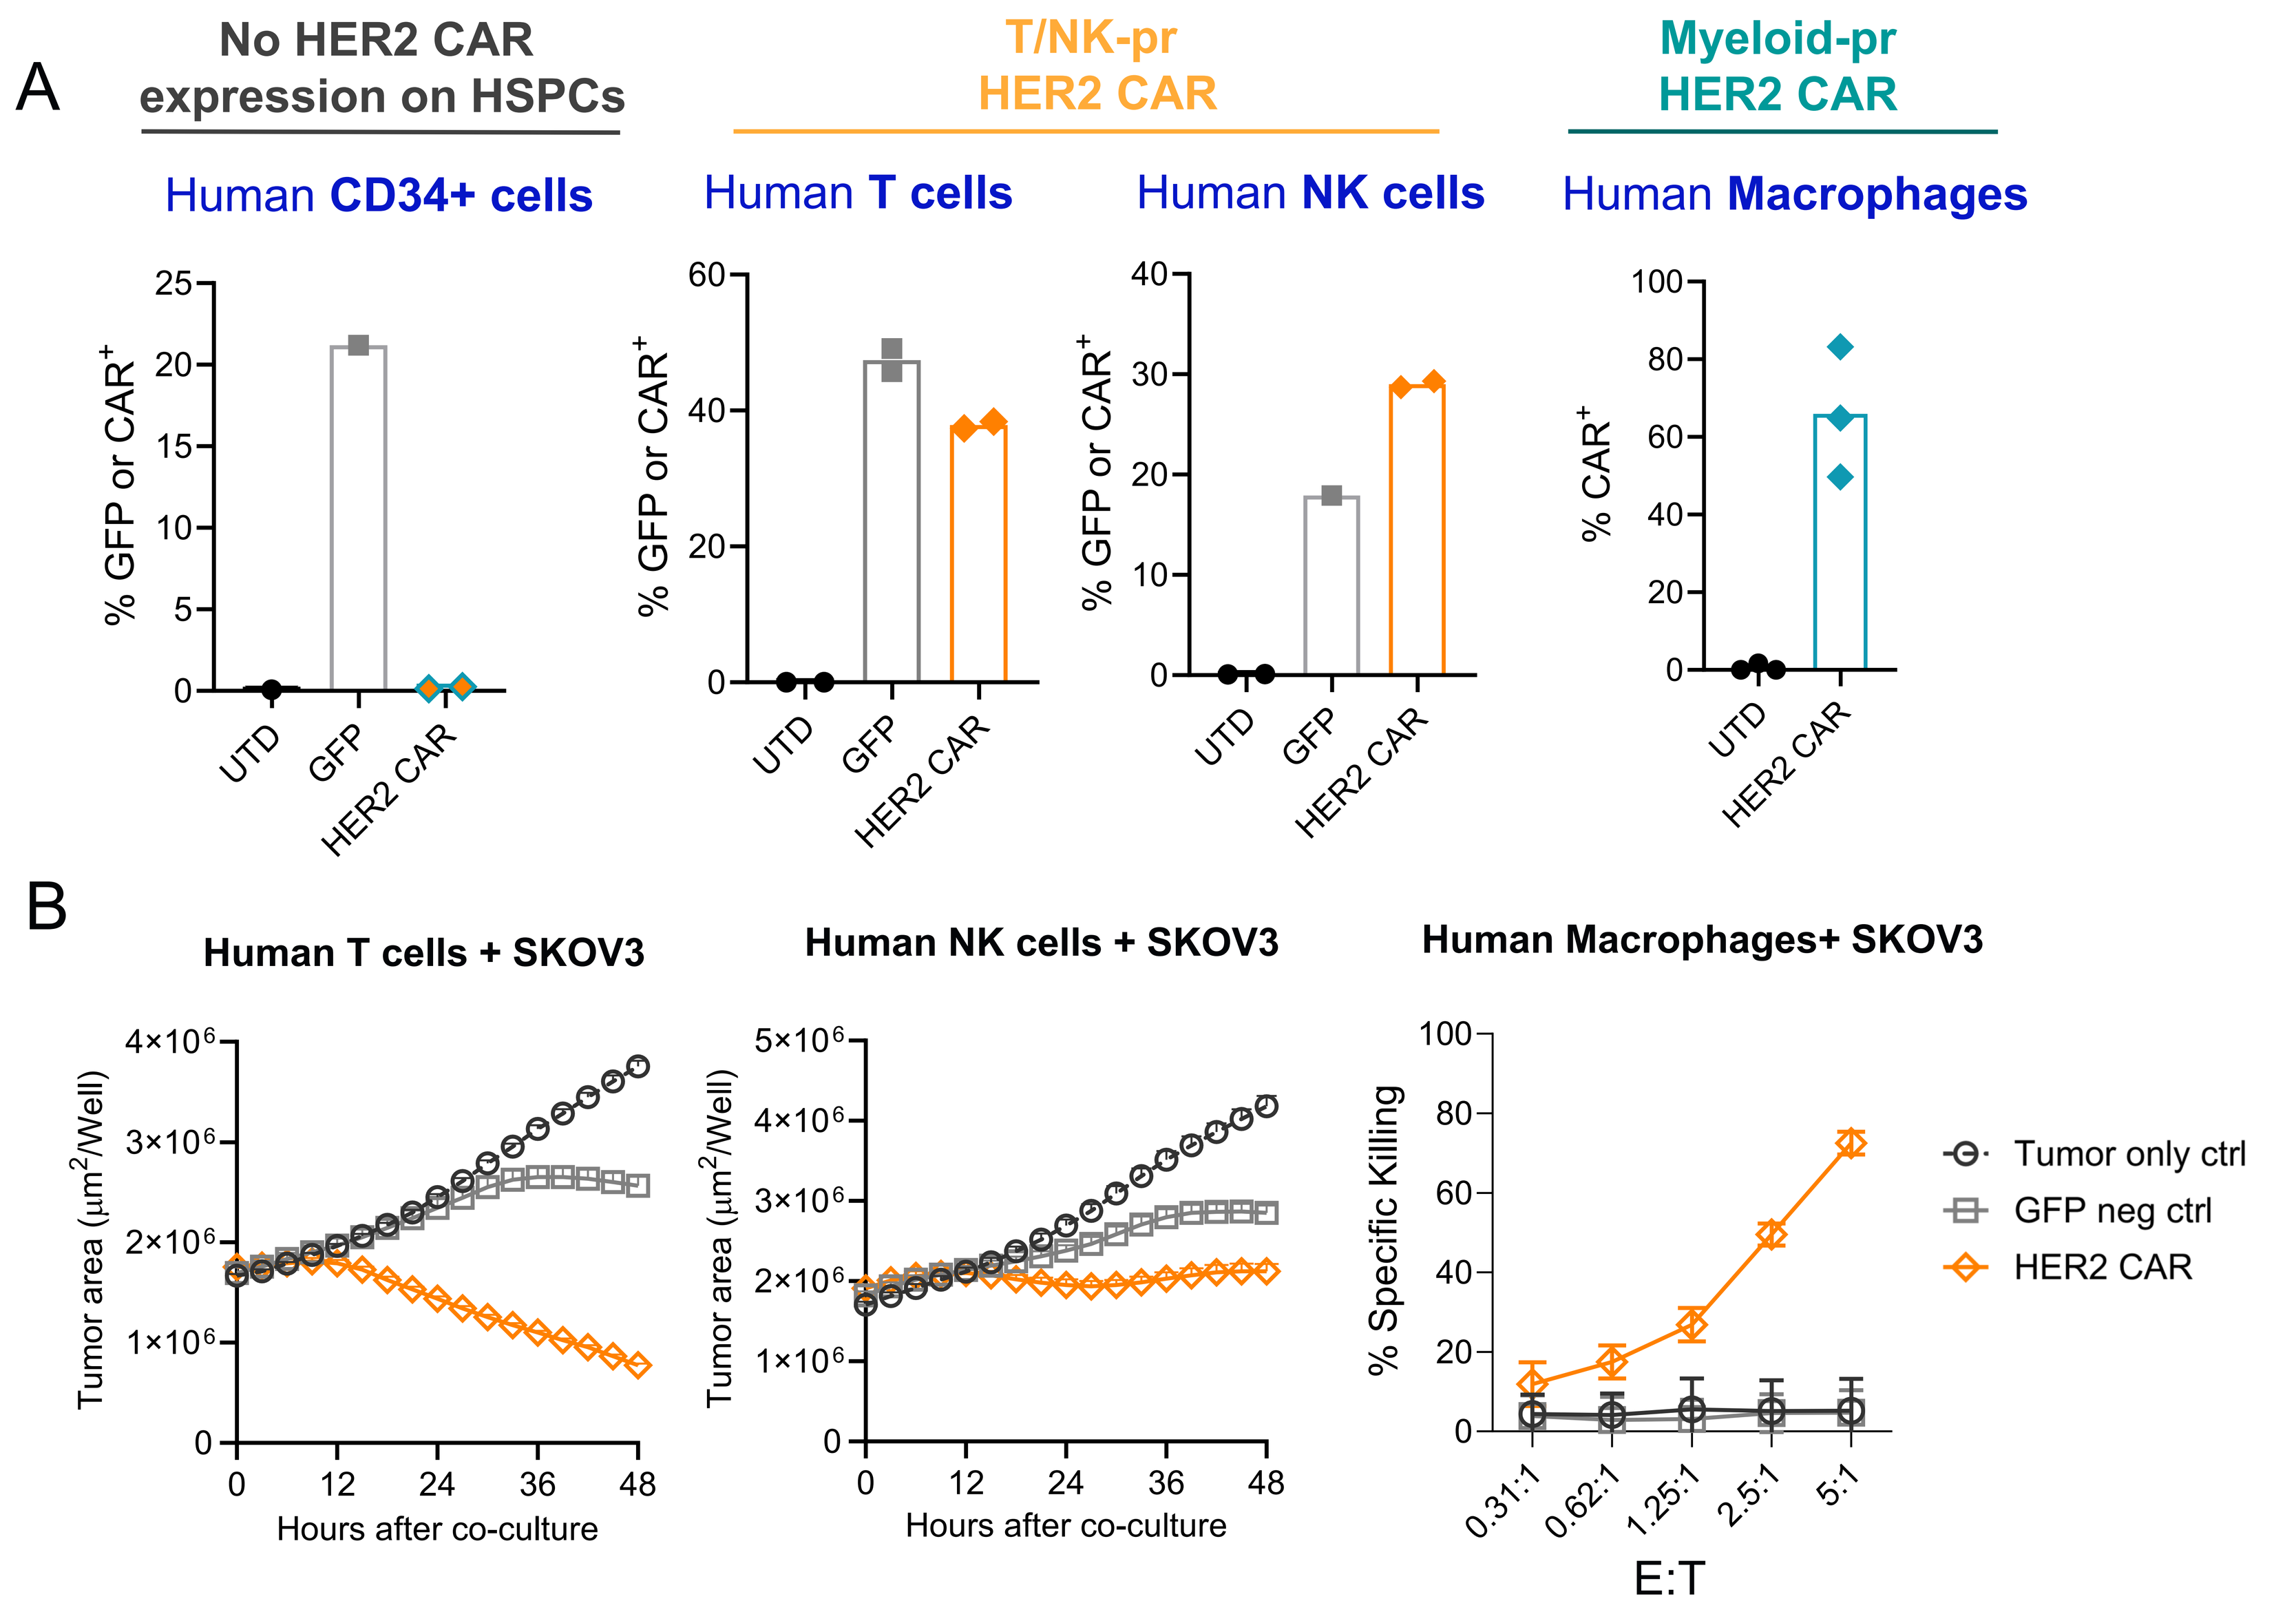
A
No HER2 CAR expression on HSPCs
T/NK-pr HER2 CAR
Myeloid-pr HER2 CAR
Human CD34+ cells
Human T cells
Human NK cells
Human Macrophages
% GFP or CAR+
% CAR+
UTD
GFP
HER2 CAR
B
Human T cells + SKOV3
Human NK cells + SKOV3
Human Macrophages+ SKOV3
Tumor area (µm²/Well)
Hours after co-culture
% Specific Killing
E:T
Tumor only ctrl
GFP neg ctrl
HER2 CAR

Figure 1. (A) *In vitro* HER2 CAR expression in human CD34+ stem cells, T cells, NK cells, and macrophages. Data represent Mean +/- SEM. **(B)** CAR-T, CAR-NK, and CAR-M from (A) were co-cultured with HER2⁺ SKOV3 tumor cells to assess cytotoxicity. Cells transduced with a VLP encoding GFP payload were used as negative control. T and NK cells were co-cultured at a 1:1 E:T ratio; CAR-M were co-cultured for 48 hours. CAR-T, CAR-NK, and CAR-M each mediate robust tumor cell killing.

Multiplexed, lineage-restricted vector generates HER2 CAR-M, NK, and T cells *in vivo*

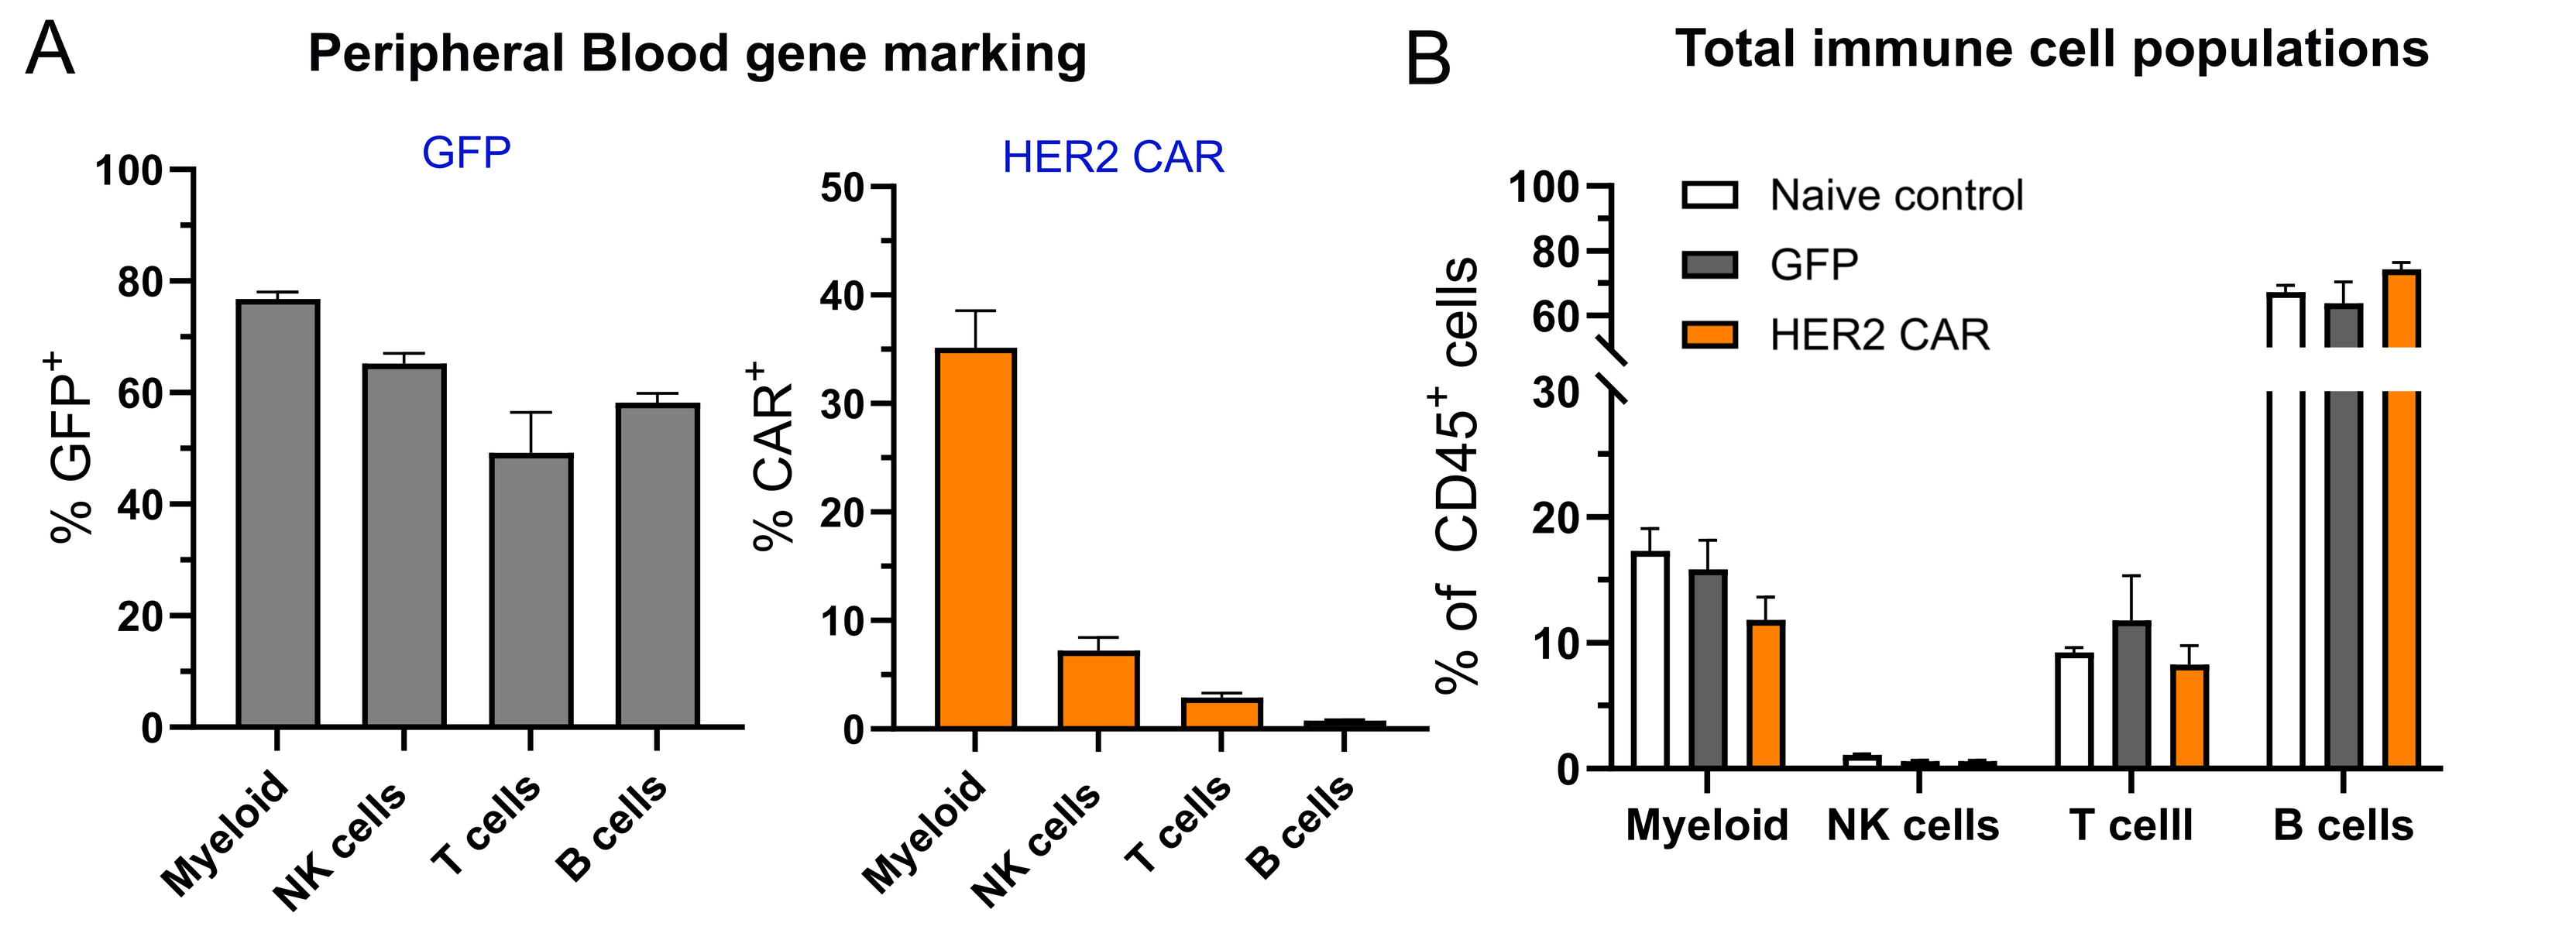
A
Peripheral Blood gene marking
GFP
HER2 CAR
% GFP+
% CAR+
Myeloid
NK cells
T cells
B cells
B
Total immune cell populations
Naive control
GFP
HER2 CAR
% of CD45+ cells
Myeloid
NK cells
T celll
B cells

Figure 2. (A) Frequency of GFP or HER2 CAR expression across mature immune cells in peripheral blood. **(B)** Frequency of HSC-derived mature immune cells of total CD45⁺ cells in peripheral blood showing immune reconstitution in mice expressing the lineage-restricted HER2 CARs is comparable as in untreated mice. N=6-8 mice per group. Data represent Mean +/-SEM.

In vivo generated, HSC-derived CAR-T cells mediate potent tumor cell killing *ex vivo*

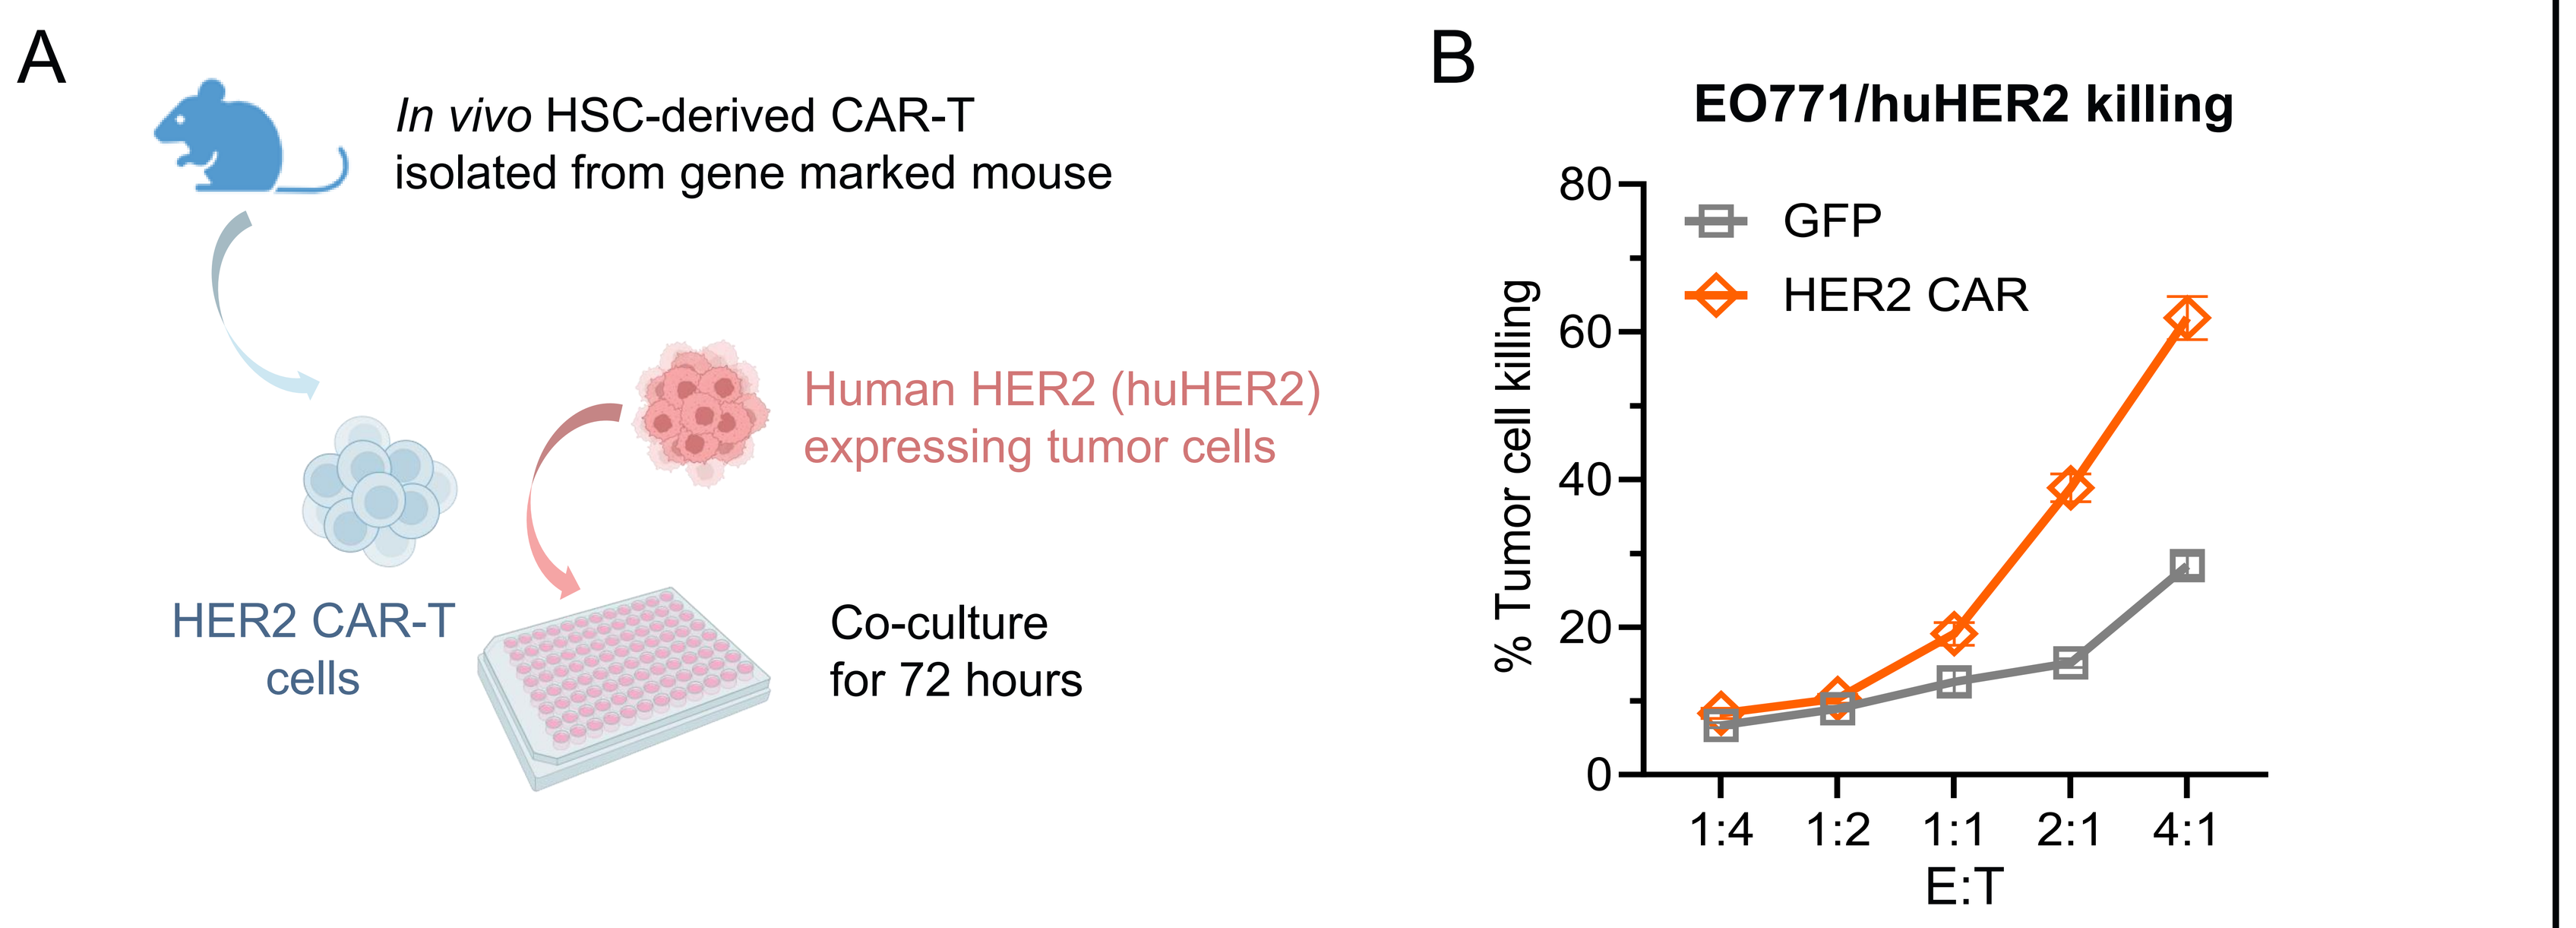
A
In vivo HSC-derived CAR-T isolated from gene marked mouse
Human HER2 (huHER2) expressing tumor cells
HER2 CAR-T cells
Co-culture for 72 hours
B
EO771/huHER2 killing
GFP
HER2 CAR
% Tumor cell killing
E:T

Figure 3. (A) *In vivo* HSC-derived CAR-T cells were isolated from gene marked mice and co-cultured with huHER2 expressing EO771 tumor cells. **(B)** Lineage-restricted, HSC-derived CAR-T cells mediate potent dose-dependent cytotoxicity. Data represent Mean +/- SEM of 4 technical replicates.

Multiplexed HER2 CAR-M, NK, & T cell therapy mediates strong tumor control *in vivo*

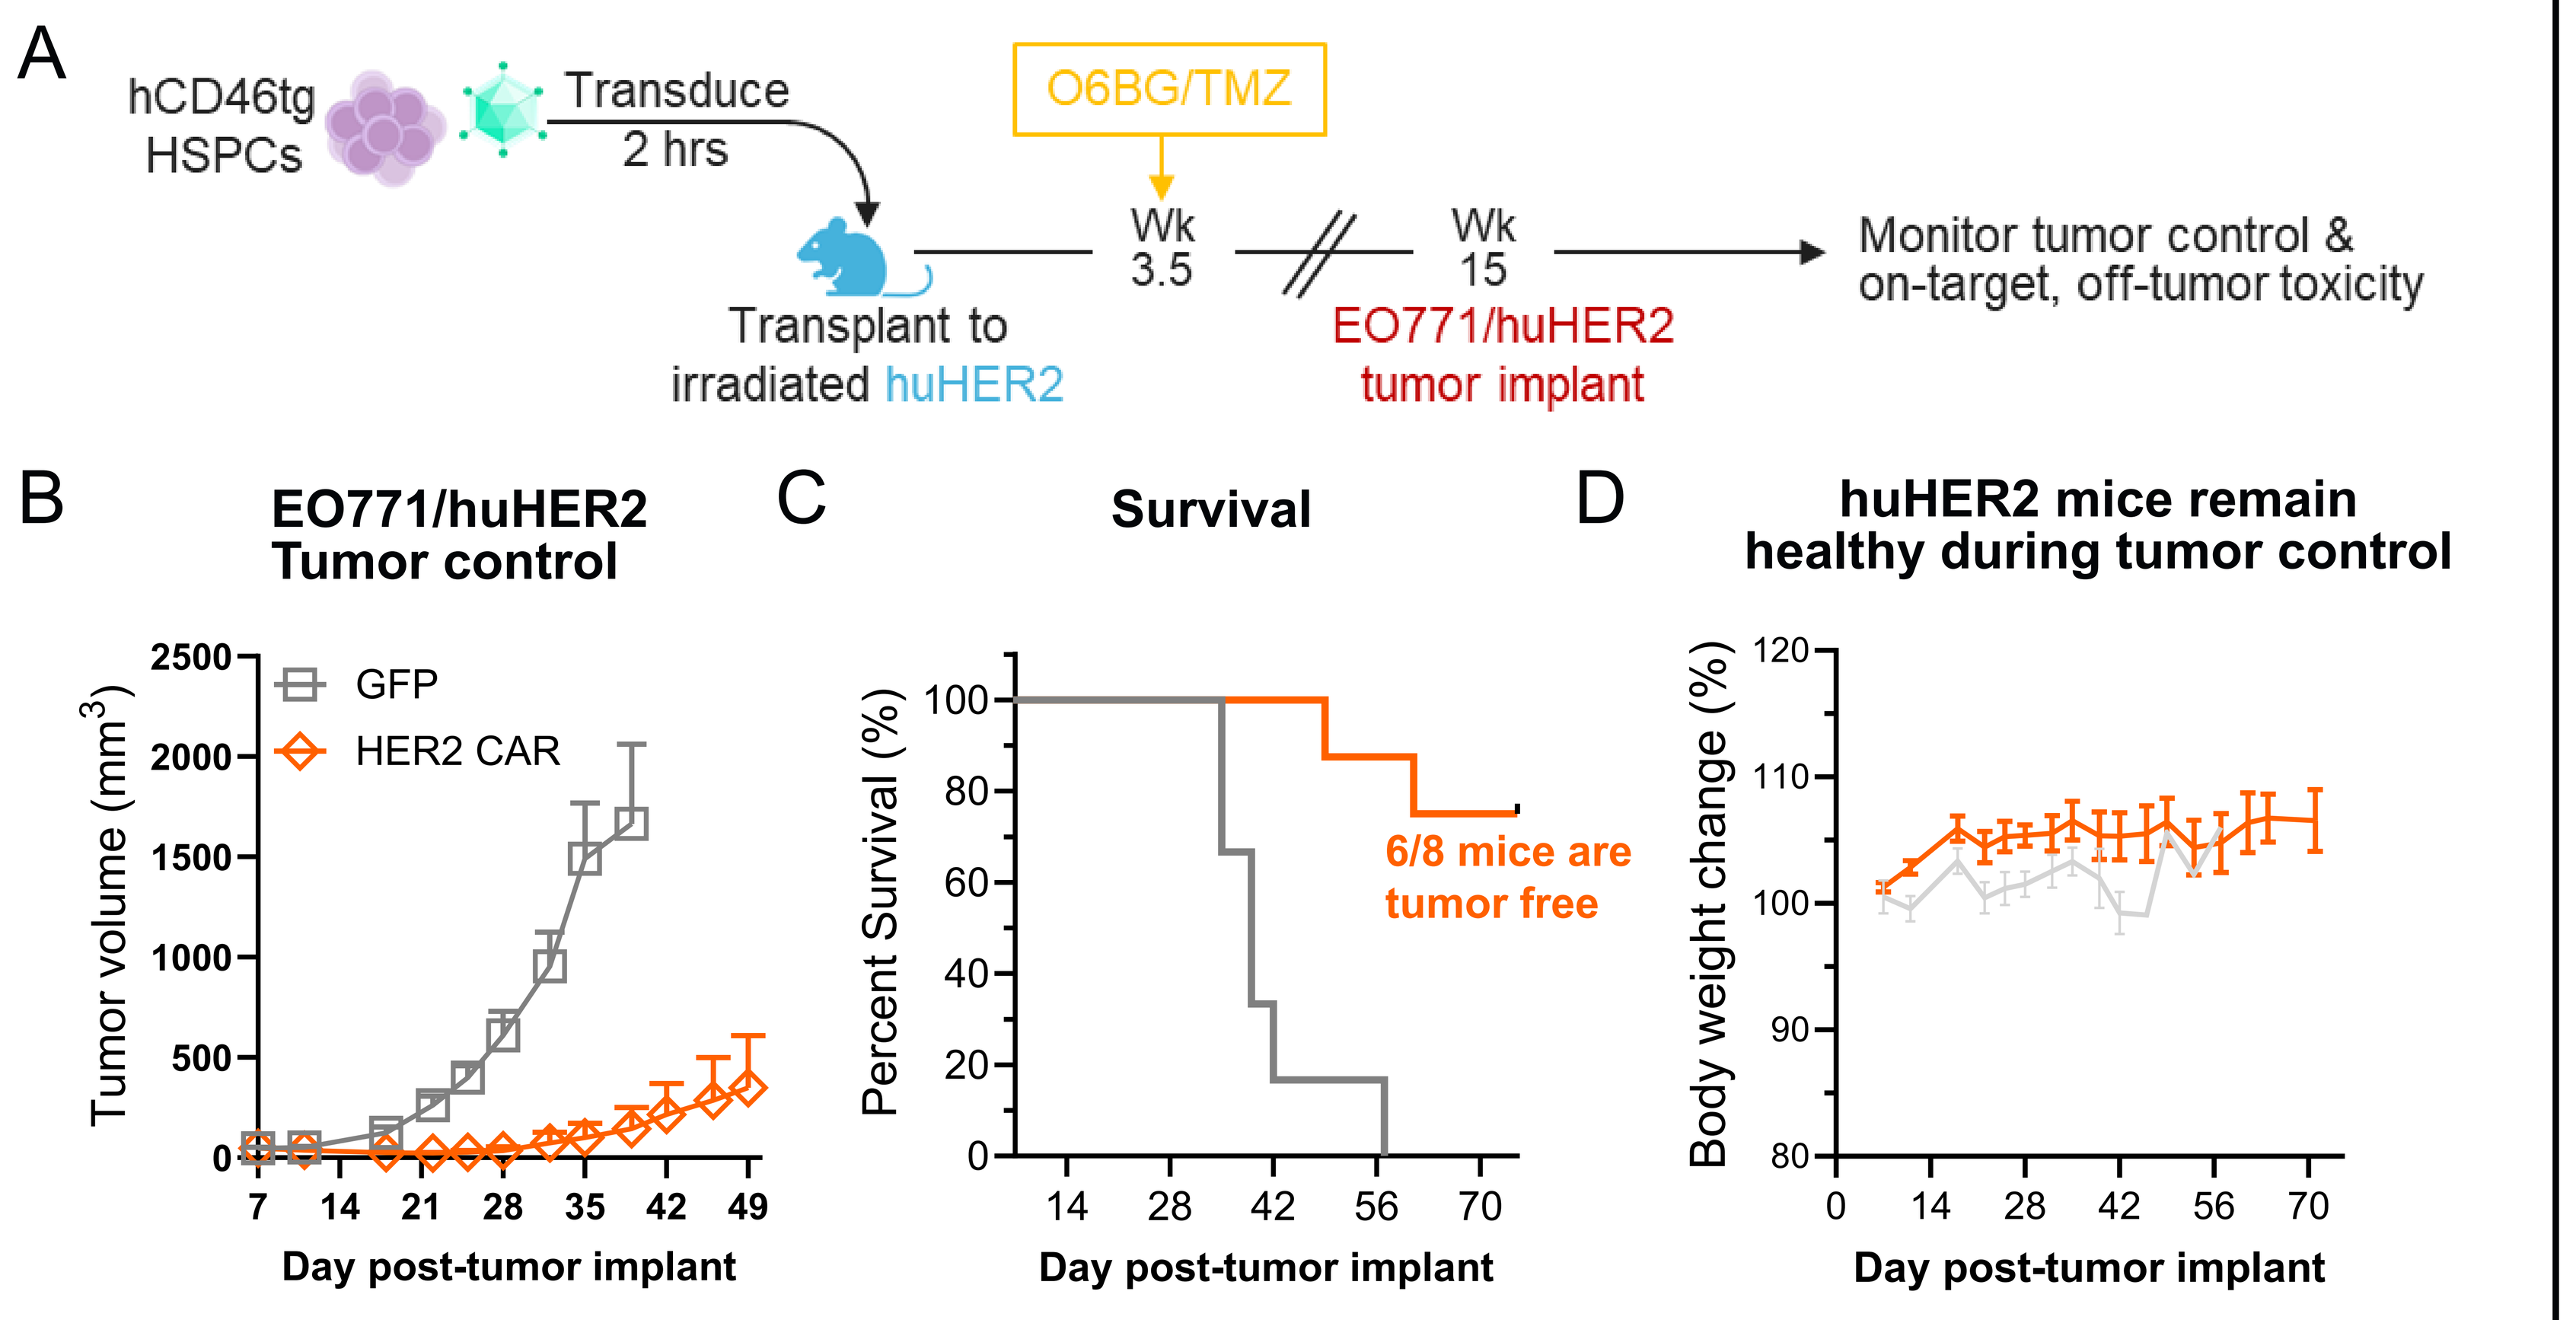
A
hCD46tg HSPCs
Transduce 2 hrs
O6BG/TMZ
Transplant to irradiated huHER2
Wk 3.5
Wk 15
EO771/huHER2 tumor implant
Monitor tumor control & on-target, off-tumor toxicity
B
EO771/huHER2 Tumor control
GFP
HER2 CAR
Tumor volume (mm³)
C
Survival
Percent Survival (%)
6/8 mice are tumor free
D
huHER2 mice remain healthy during tumor control
Body weight change (%)
Day post-tumor implant

Figure 4. (A) Schematic of the tumor control study in humanized HER2 mice receiving *ex vivo* transduced HSPCs from hCD46tg mice. EO771/huHER2 cells were implanted at week 15 post-transplant. Tumor growth curve, survival, and body weight changes were captured throughout the study. **(B)** Tumor volume was measured by caliper twice per week. **(C)** Mice were euthanized when tumors reached 2000 mm³, and fatality was recorded for survival analysis. **(D)** HER2 CAR-expressing mice maintain body weight during and following tumor clearance, suggesting HER2 CAR-M, NK, and T cells do not mediate on-target, off-tumor toxicity in this model. N=6-8 per group. Data represent Mean +/-SEM.

Conclusions

- Lineage-specific promoters direct CAR expression to discreet mature immune cells, enabling **regulated expression of multiplexed therapeutic payloads**
- *In vivo* multiplexed, lineage-restricted vector generates **HSC-derived CAR-M, CAR-NK, & CAR-T** cells and mediates **robust anti-tumor efficacy**
- Preparing to advance development candidate **EN-002 to clinical studies in 2027**

Related talks

Late-Breaking Abstract: First in vivo hematopoietic stem cell (HSC) gene addition clinical trial: Initial results from EN-374-101 in X-linked chronic granulomatous disease (X-CGD). Friday, May 15, 11:07AM – 11:33AM ET

Oral Abstract: Discovery and development of engineered neutralizing antibody-evading helper-dependent adenovirus capsids as candidates for in vivo gene therapy. Wednesday, May 13, 11:15AM – 11:30AM ET

Created with BioRender.com

ensoma

CONFIDENTIAL